Multivariant Analysis to Decipher the Human Pancreatic Cancer Proteome to Identify Novel Biomarkers and Therapeutic Targets

Henry C. H. Law, Dragana Lagundzin, Zachery S. Wagner, Nicholas T. Woods
Eppley Institute for Research in Cancer, University of Nebraska Medical Center

UNIVERSITY OF NEBRASKA MEDICAL CENTER

Abstract Number: 5647

Abstract

The microenvironment of a tumor plays a vital role in its development and progression. We hypothesized that the pancreatic ductal adenocarcinoma (PDAC) tumor microenvironment could elucidate proteins associated with patient survival.

This project utilized a quantitative proteomics approach designed to:

1. Identify differentially expressed proteins in the tumor microenvironment of PDAC liver metastases that exhibited different levels of cancer cell differentiation;
2. Ascertain proteins associated with PDAC patient survival to build a predictive model for prognosis; and
3. Identify potential therapeutic targets to promote patient survival.

The tissue samples were preserved within 1-3 hours to avoid post-mortem protein degradation. The tissues were divided into 7 batches and were lysed, digested and differentially labeled with isobaric Tandem Mass Tags. The tagged peptides were mixed, fractionated with C18 spin columns and analyzed by an Orbitrap Fusion Lumos LC-MS/MS system. 30811 peptides and 3960 proteins were identified from the analysis. 1842 of them were quantified and 917 proteins were submitted for principal component analysis.

The tandem mass tag (TMT) methodology

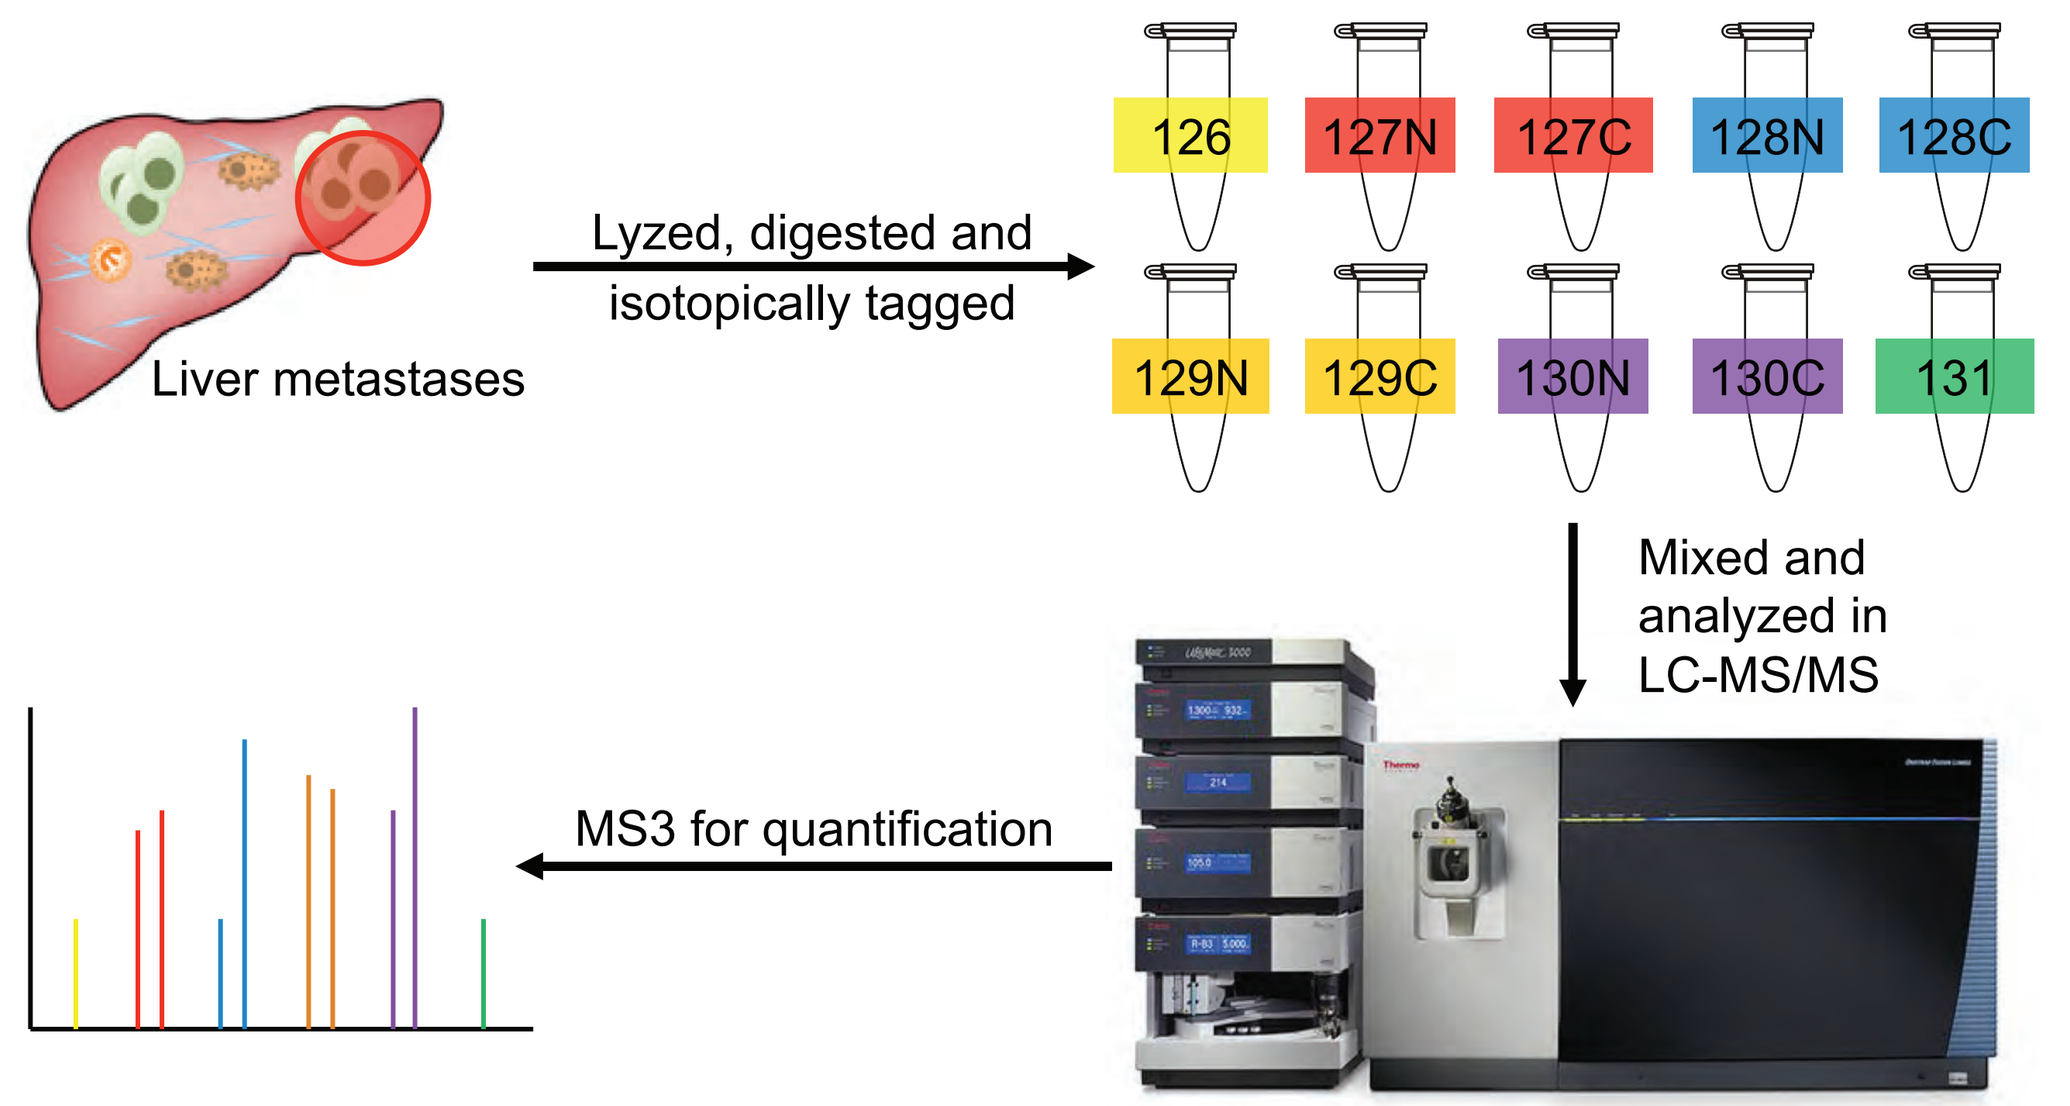
Liver metastases
Lyzed, digested and isotopically tagged
126
127N
127C
128N
128C
129N
129C
130N
130C
131
Mixed and analyzed in LC-MS/MS
MS3 for quantification

Figure 1. The proteomic analysis workflow for the rapid autopsy samples.
The RAP sample proteome from 60 individuals was divided into 7 batches and quantified by the TMT methodology. Briefly, the proteins were first extracted from the liver metastases tissue and were digested into peptides. The peptides were then differentially tagged with different stable isotopes (127N-131) for quantification in the shotgun proteomic analysis. The 126 channel was reserved for the master mix of the 60 samples and served as a common reference across the 7 batches of samples. The tagged peptides were then mixed and analyzed with the Orbitrap Fusion Lumos. The abundance of the proteins in each of the samples was estimated by the reporter ion ratio observed in the MS3 spectra of the corresponding peptides in each of the protein observed.

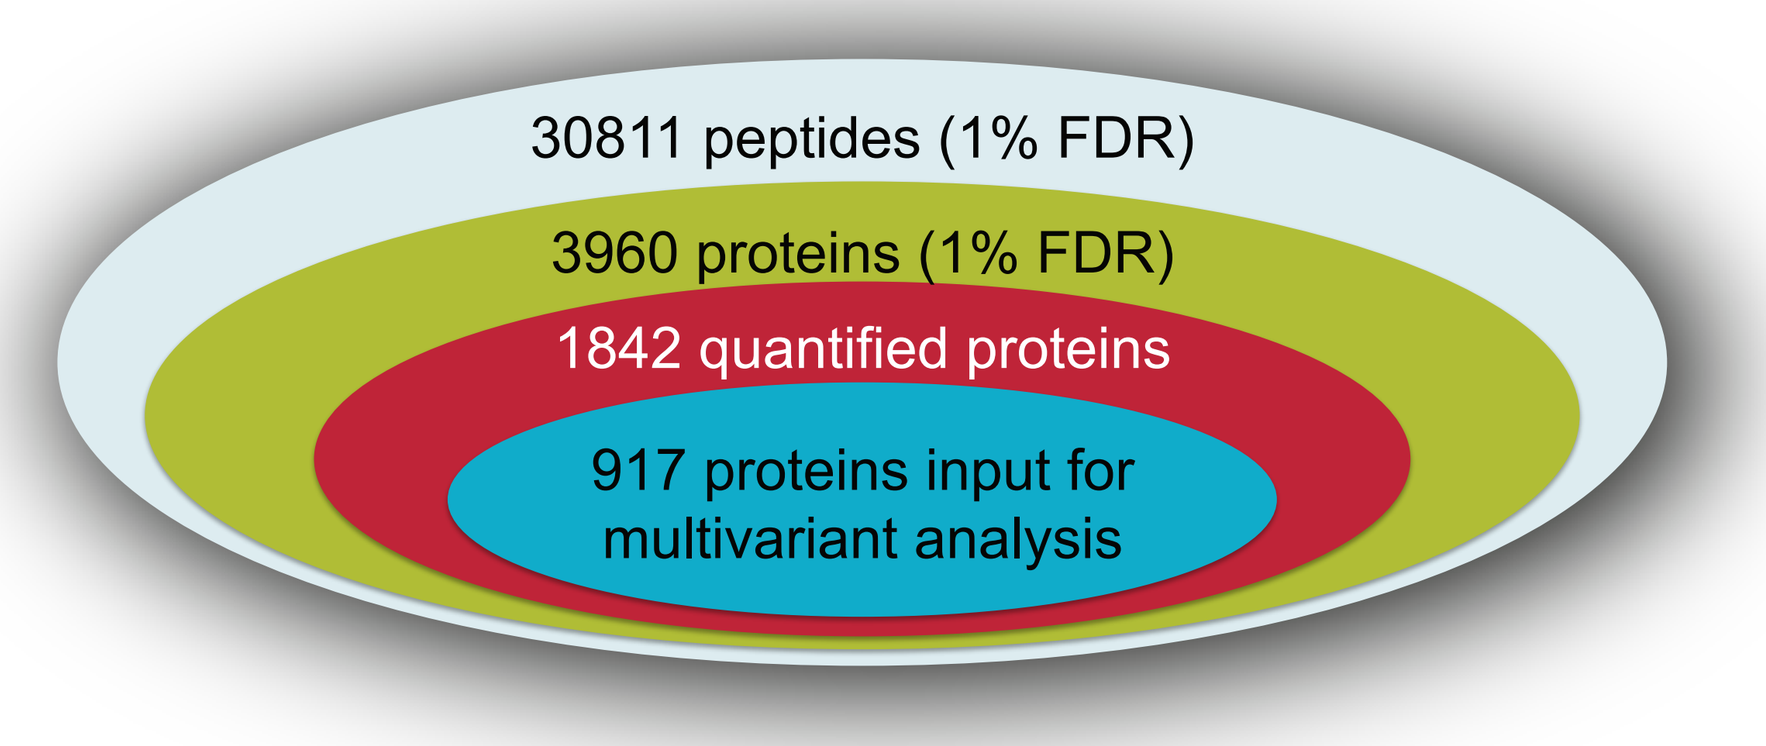
30811 peptides (1% FDR)
3960 proteins (1% FDR)
1842 quantified proteins
917 proteins input for multivariant analysis

Figure 2. An overview of the proteomics data.
Quantified proteins are defined as proteins with at least 5 spectra from the database search. Proteins quantified in more than 80% of the samples are included in the multivariant analysis.

Malignant versus Normal Adjacent Tissue

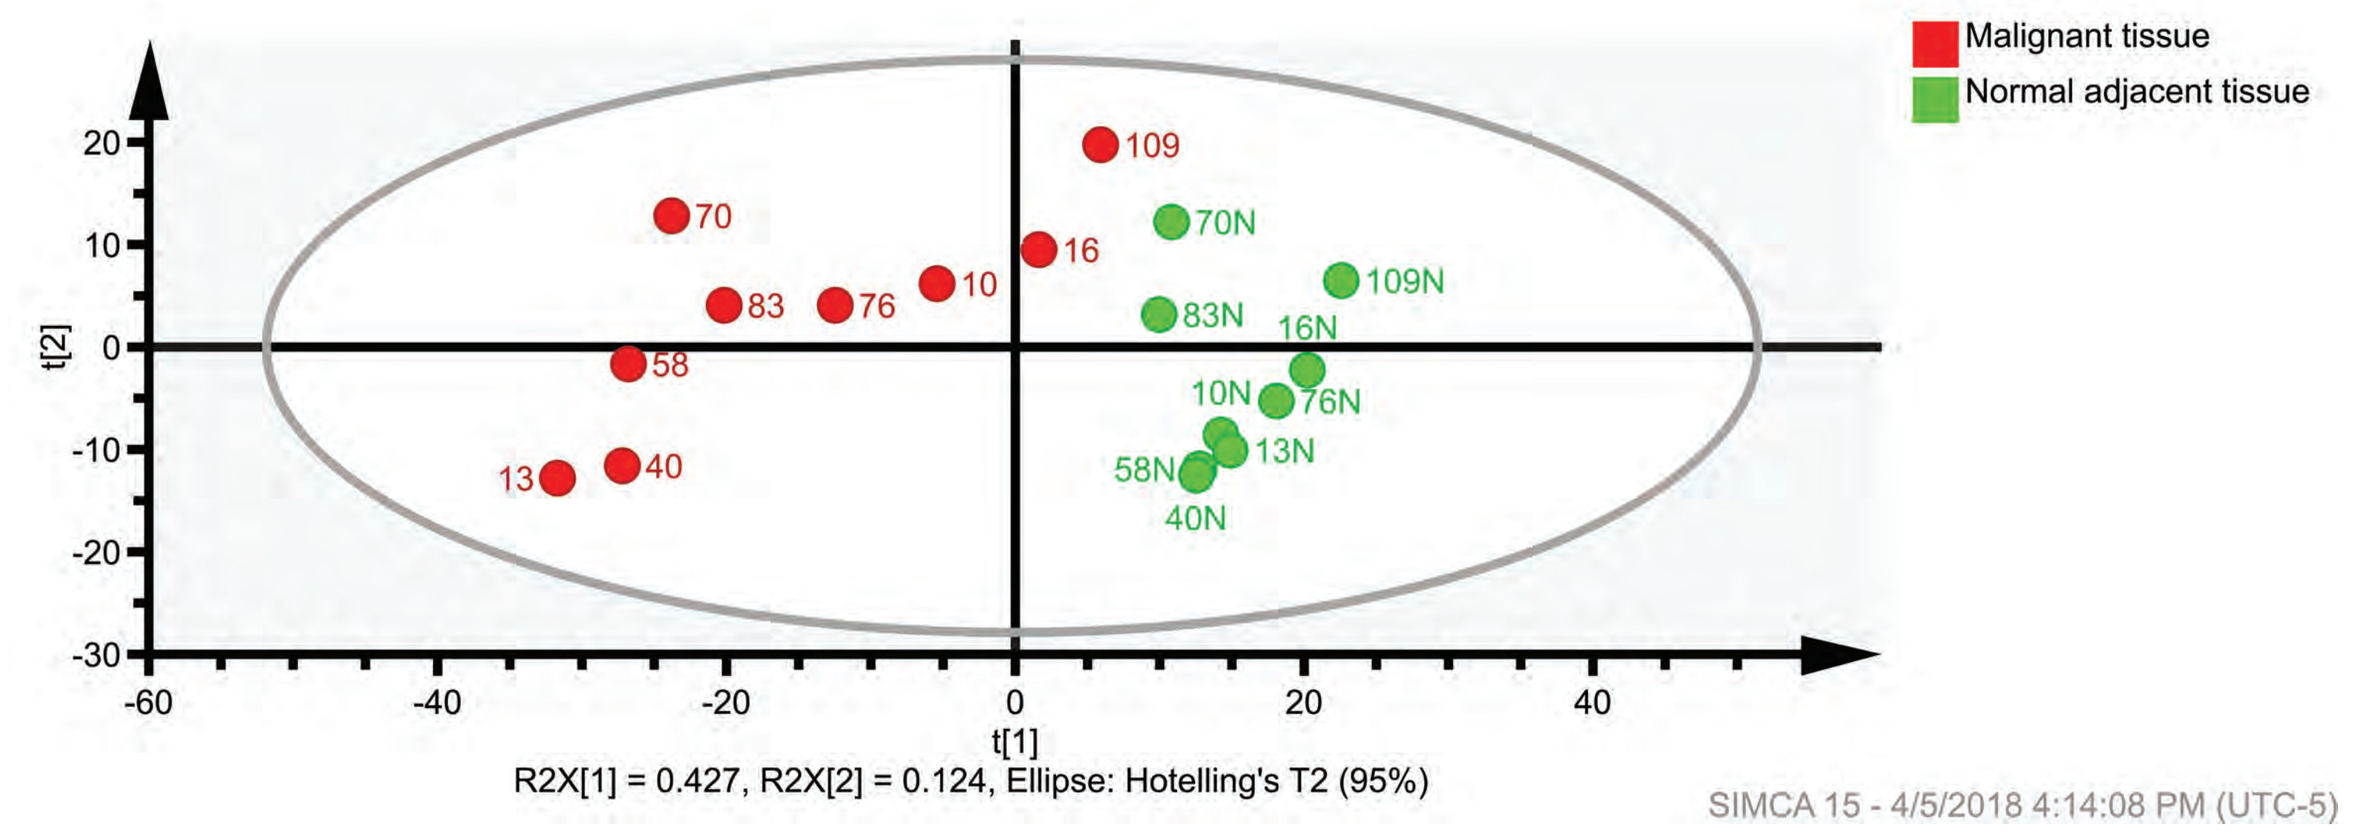
Malignant tissue
Normal adjacent tissue
t[1]
t[2]
R2X[1] = 0.427, R2X[2] = 0.124, Ellipse: Hotelling's T2 (95%)
SIMCA 15 - 4/5/2018 4:14:08 PM (UTC-5)

Figure 3. Score plot of the malignant versus the normal adjacent tissue proteome in the Principal Component Analysis (PCA).
This analysis aimed to check if different tissue types can be discerned in Principal Compoenet Analysis. The proteome of 9 malignant liver metastases tissues and their corresponding normal adjacent tissues were submitted to Principal Component Analysis. The vectors representing the malignant tissues and the normal adjacent tissues aligned according the tissue type on the score plot. It suggested that the Principal Component Analysis provided sufficient sensitivity of different tissue types in the tumor, and thus can be used for phenotyping PDAC tissues on the next stage. The numbers on the plot indicates the patient number as registered in the Rapid Autopsy Program. The suffix "N" signified the normal adjacent tissues. ($R^2X(cum) = 0.7078$; $Q^2(cum) = 0.4006$)

PDAC Phenotyping with Multivariant Analysis (MVA)

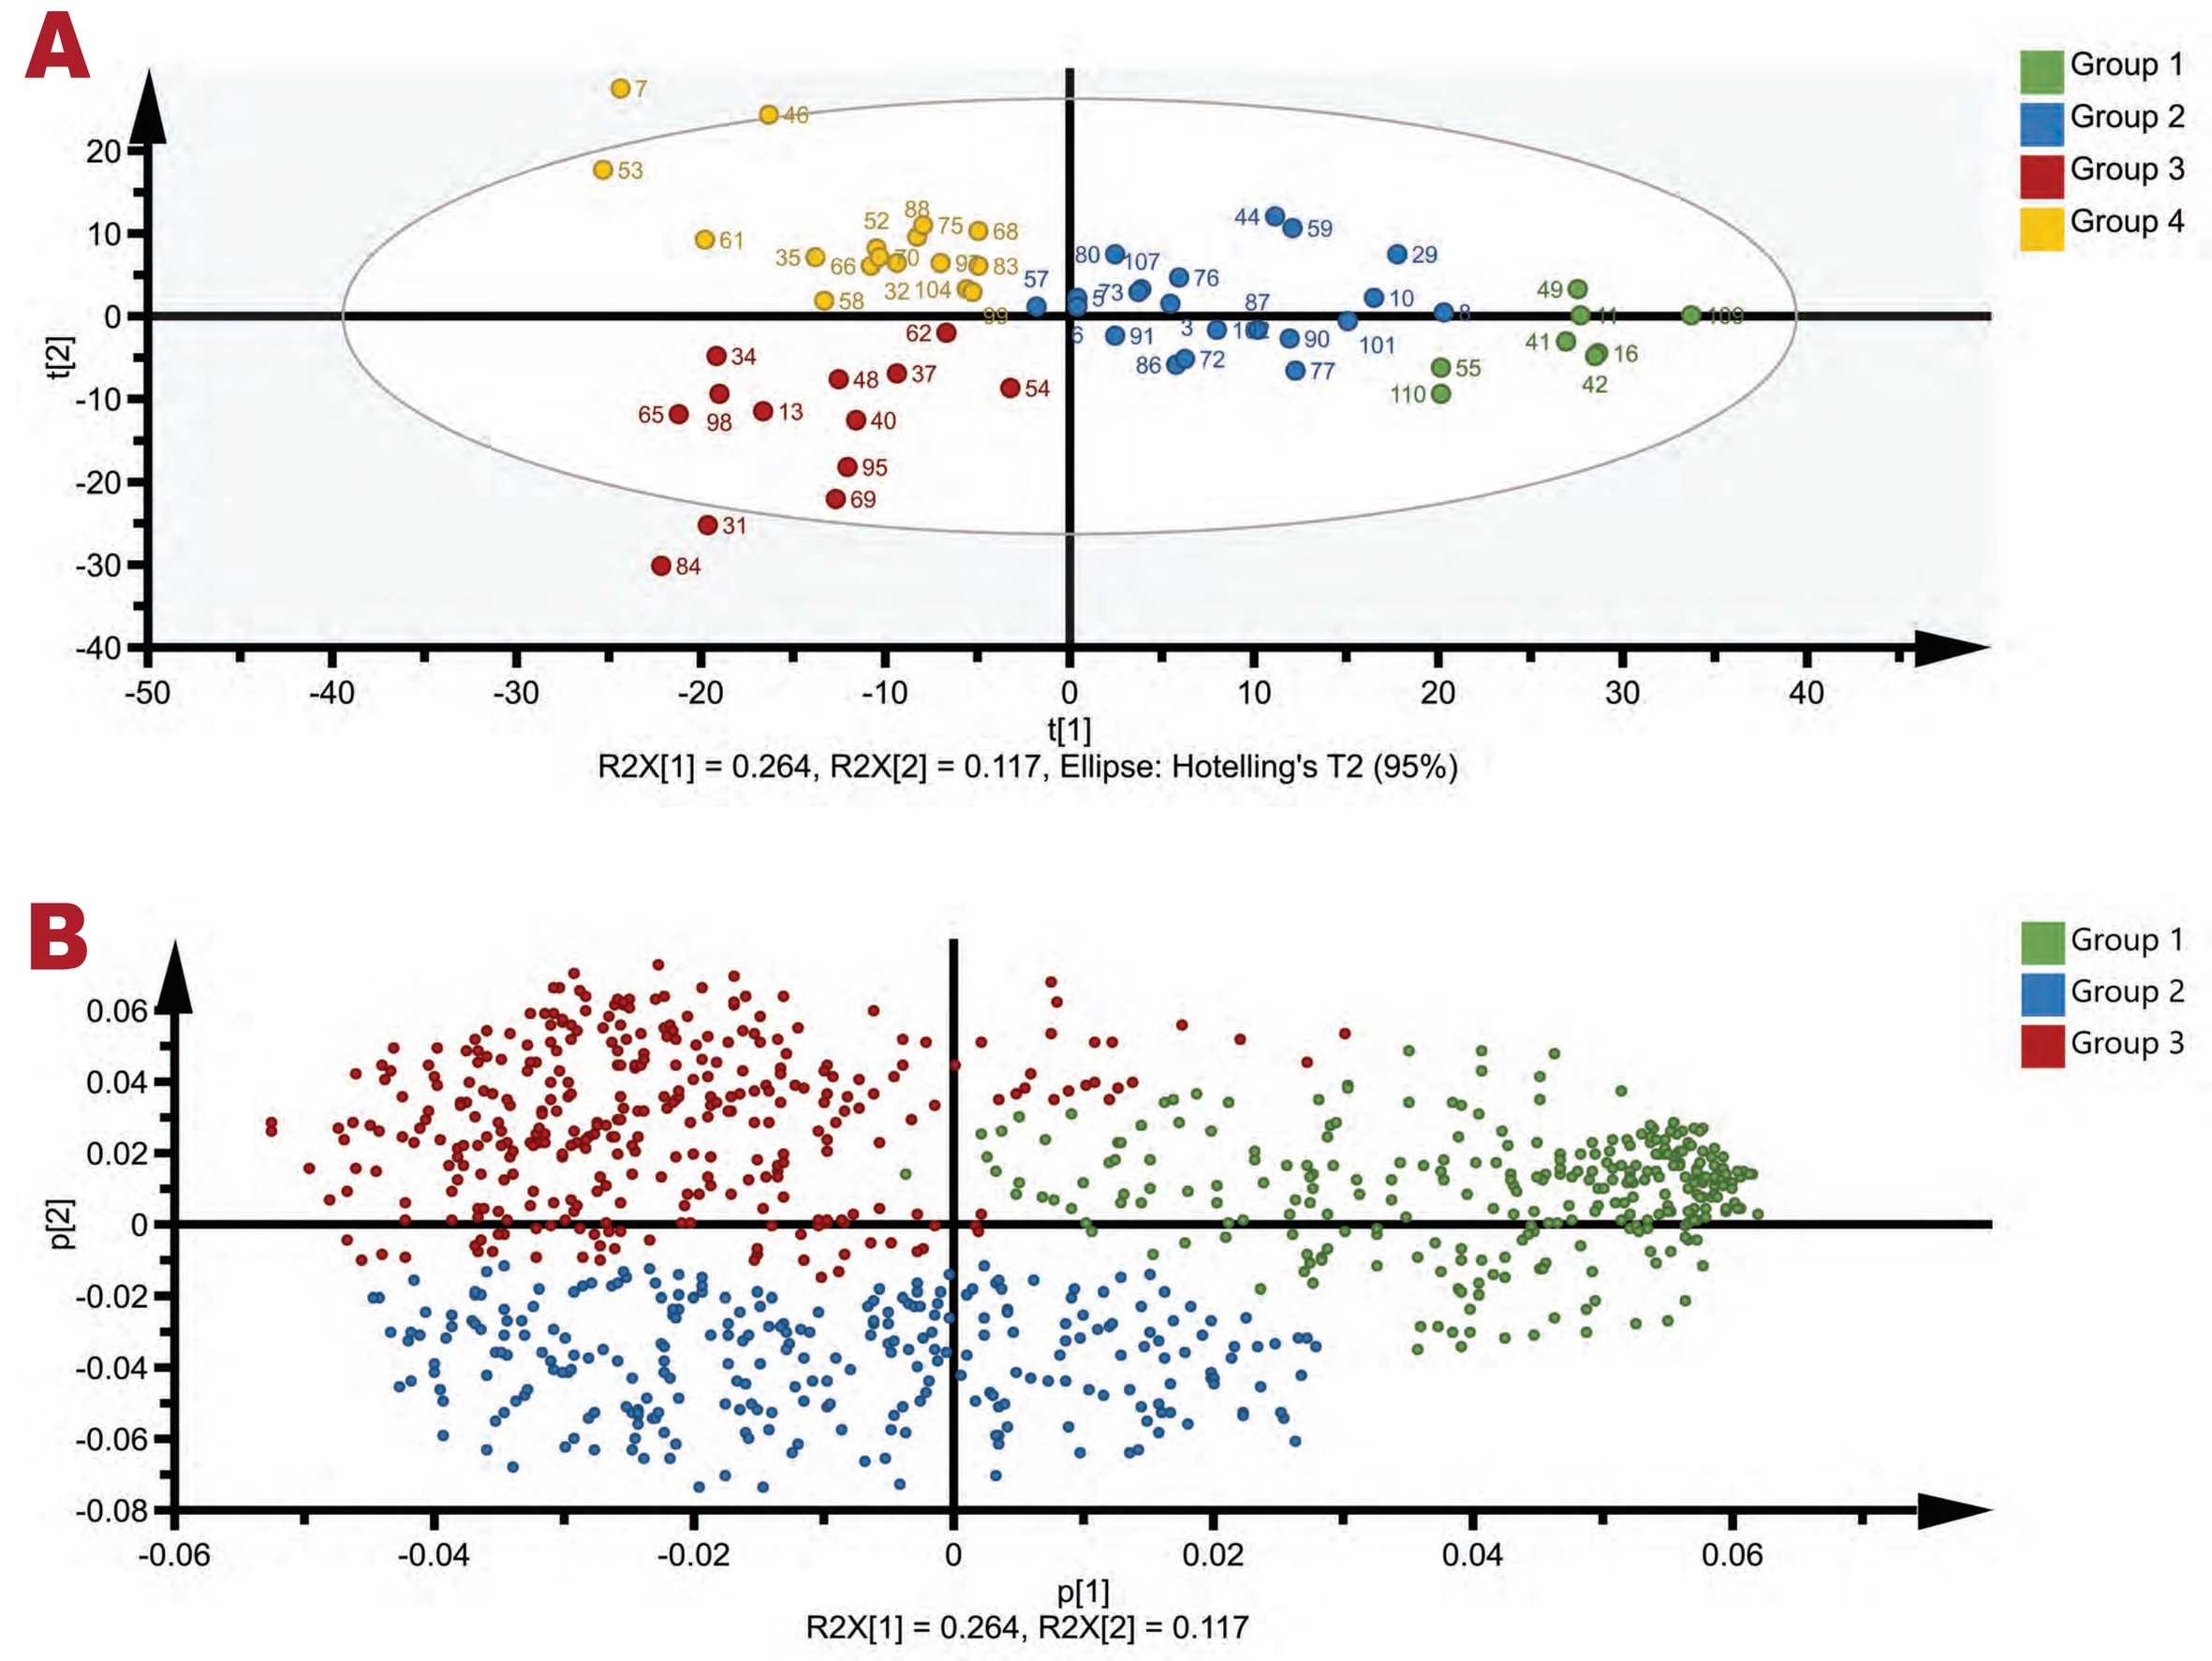
A
Group 1
Group 2
Group 3
Group 4
t[1]
t[2]
R2X[1] = 0.264, R2X[2] = 0.117, Ellipse: Hotelling's T2 (95%)
B
Group 1
Group 2
Group 3
p[1]
p[2]
R2X[1] = 0.264, R2X[2] = 0.117

Figure 3. The (A) score plot and (B) loading plot of the PDAC proteome Principle Component Analysis.
917 commonly quantified prtoeins across the 7 batches of samples were input to the Principle Component Analysis to look similar expression patterns across the proteome and the samples. Samples with similar protein expression patterns, and proteins with similar expression patterns were plot close to each other on the (A) score plot and (B) loading plot, respectively. Base on the geometrical spacing between the samples on the score plot, the 59 samples were grouped into 4 major subtypes. Proteins driving the separation of the subtypes can be found in the corresponding quadrant on the loading plot. For instance, Group 1 on the score plot was driven by the proteins on the right hand side, labelled in green on the loading plot. The biological significance for each of the subtypes was investigate in the subsequant analysis. ($R^2X(cum) = 0.692$; $Q^2(cum) = 0.412$)

Characterization of PDAC Phenotypes

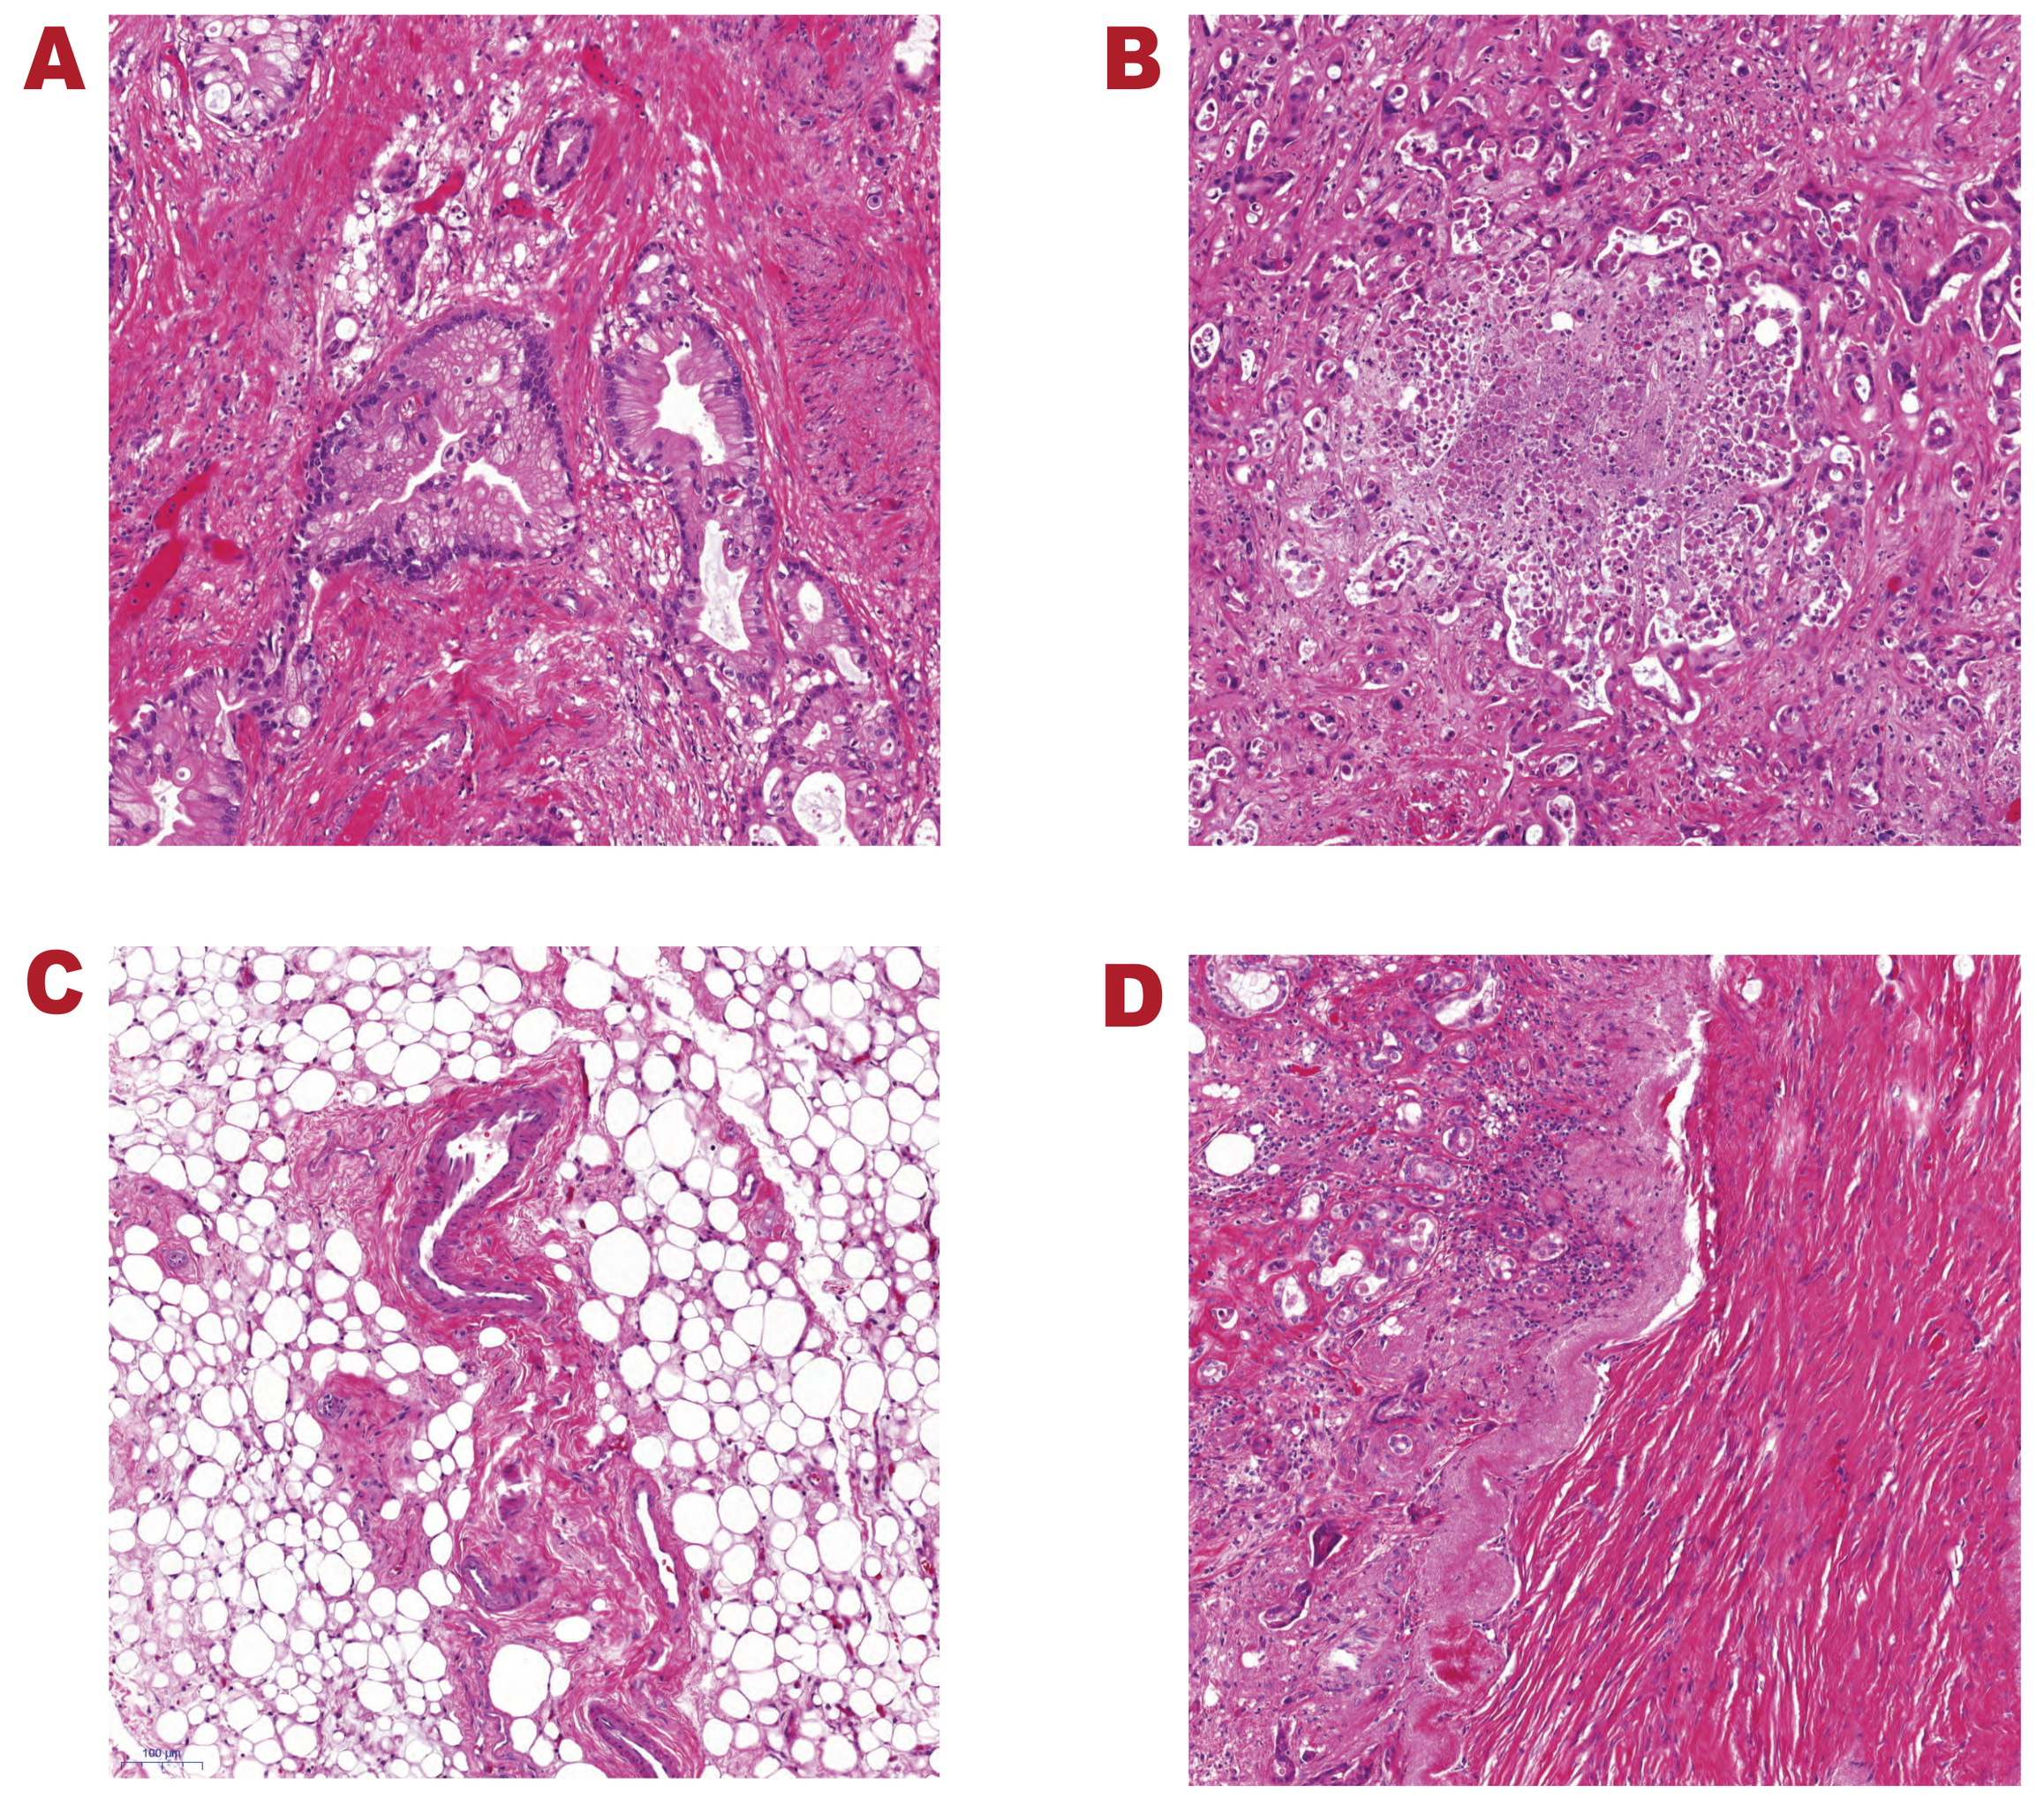
A
B
C
D

Figure 3. Representative tissue sections for each of the subtype.
Figure 3A-D showed the representative tissue section for Group 1 - 4 respectively.

Biological Process	Group 1 No. of proteins	Group 1 Fold enrichment	Group 2 No. of proteins	Group 2 Fold enrichment	Group 3 No. of proteins	Group 3 Fold enrichment
Fatty acid beta-oxidation	16	**38.23**	0	< 0.2	0	< 0.2
Tricarboxylic acid cycle	8	**22.06**	3	8.72	3	7.15
Cellular amino acid catabolic process	15	**19.2**	1	1.35	2	2.21
Gluconeogenesis	5	**17.07**	1	3.6	2	5.9
Cellular component organization	8	0.38	45	**2.27**	65	**2.69**
Cellular component organization or biogenesis	8	**0.34**	66	**2.98**	65	**2.41**
Protein complex assembly	1	0.45	17	**7.98**	4	1.54
Protein complex biogenesis	1	0.44	17	**7.93**	4	1.53
Cellular component morphogenesis	4	0.65	8	1.37	37	**5.21**
Macrophage activation	1	0.62	0	< 0.2	14	**7.54**
Cell-matrix adhesion	0	< 0.2	0	< 0.2	8	**7.18**

Table 1. Gene ontology analysis of the clustered proteins in the PCA analysis.
The clustered proteins were submitted to PANTHER for gene ontology terms enrichment analysis. The fold enrichments with p-value <0.05 were bold; the directionalities were indicated in yellow (up) and blue (down), respectively.

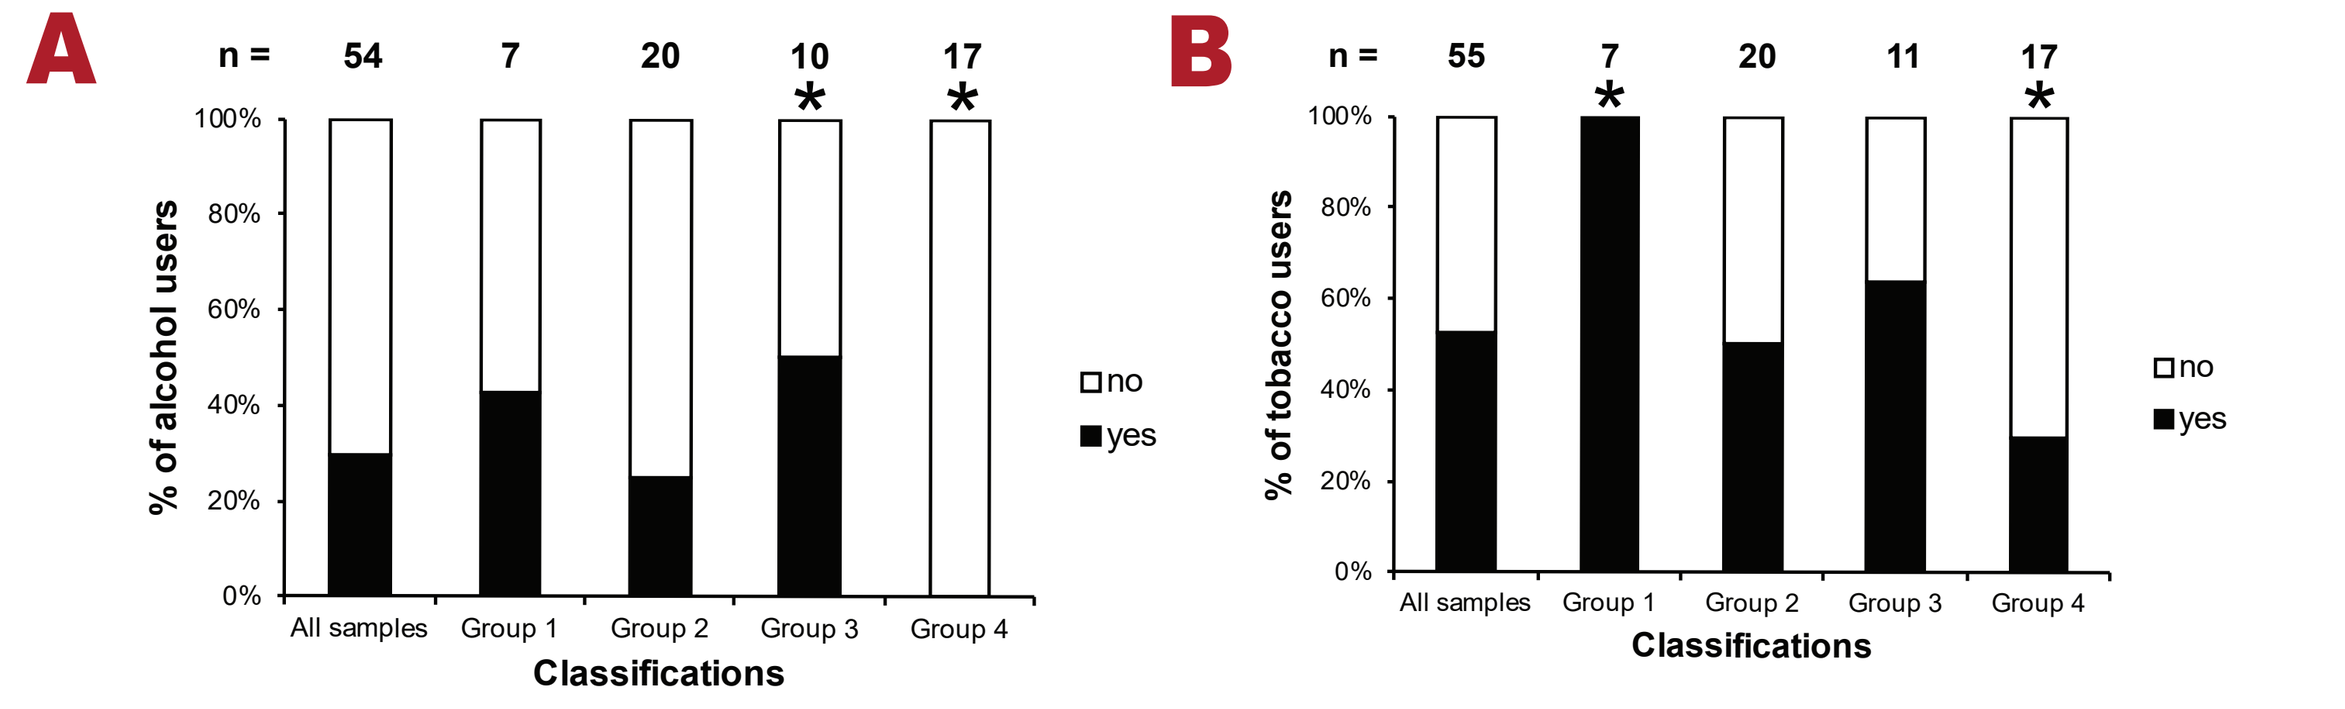
A
n = 54 7 20 10 17
% of alcohol users
no
yes
All samples
Group 1
Group 2
Group 3
Group 4
Classifications
B
n = 55 7 20 11 17
% of tobacco users
no
yes
All samples
Group 1
Group 2
Group 3
Group 4
Classifications

Figure 4. Association between (A) alcohol and (B) tobacco usage and PDAC subtype.
Alcohol and tobacco usage were found associated with the PDAC subtype identified in the proteomeics data. Asterisks(*) signified a p-value <0.05 in hypogeometric test.

Towards a Clinical Subtyping Scheme

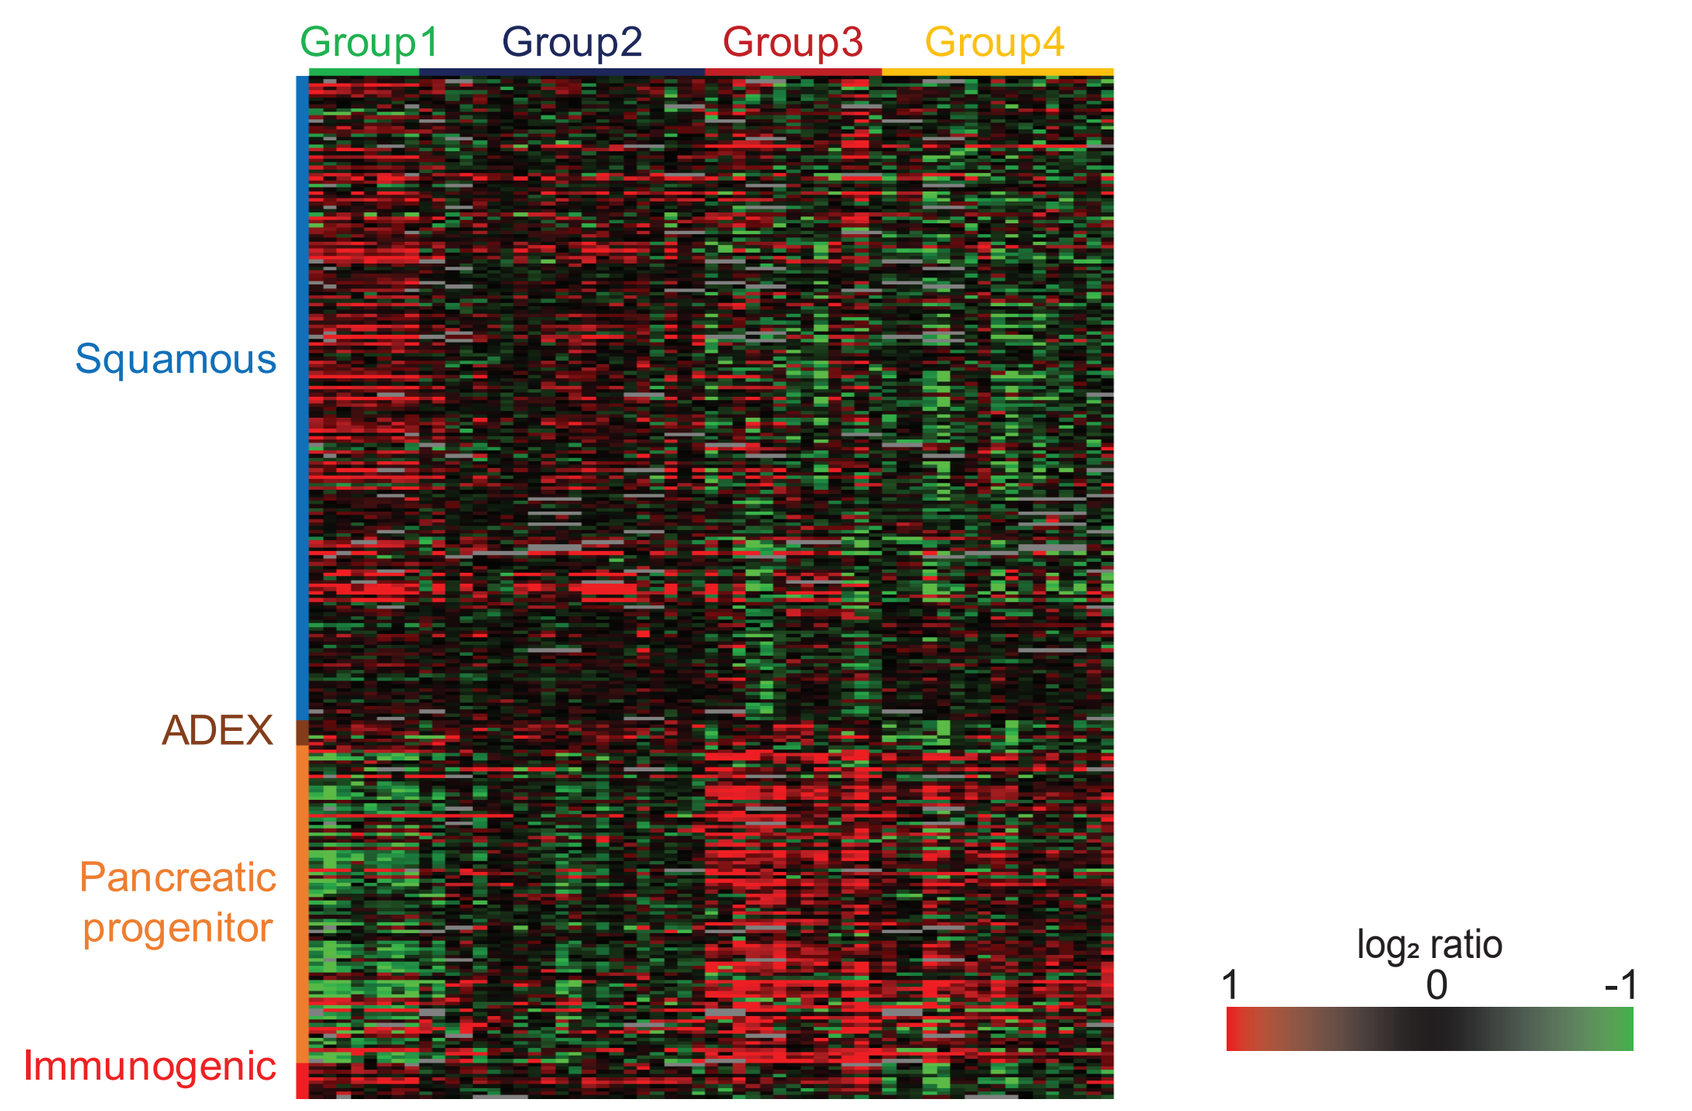
Group1
Group2
Group3
Group4
Squamous
ADEX
Pancreatic progenitor
Immunogenic
log₂ ratio
1
0
-1

Figure 5. Comparison of genome-based and proteomic based subtyping scheme
The characteristic gene clusters from the genome-based subtyping scheme devised by Bailey *et al* (2016) were extracted and compared with the current proteome data. The x- and y-axis represent the sample and the genes, repectively. The green and red in the pixels signified the up- and down-regulation of the gene, respectively. The protein expression of these gene clusters generally correlated with the subtyping scheme. However, the proteomic subtyping scheme may further differentiate some of the subtypes in the Bailey *et al* scheme.

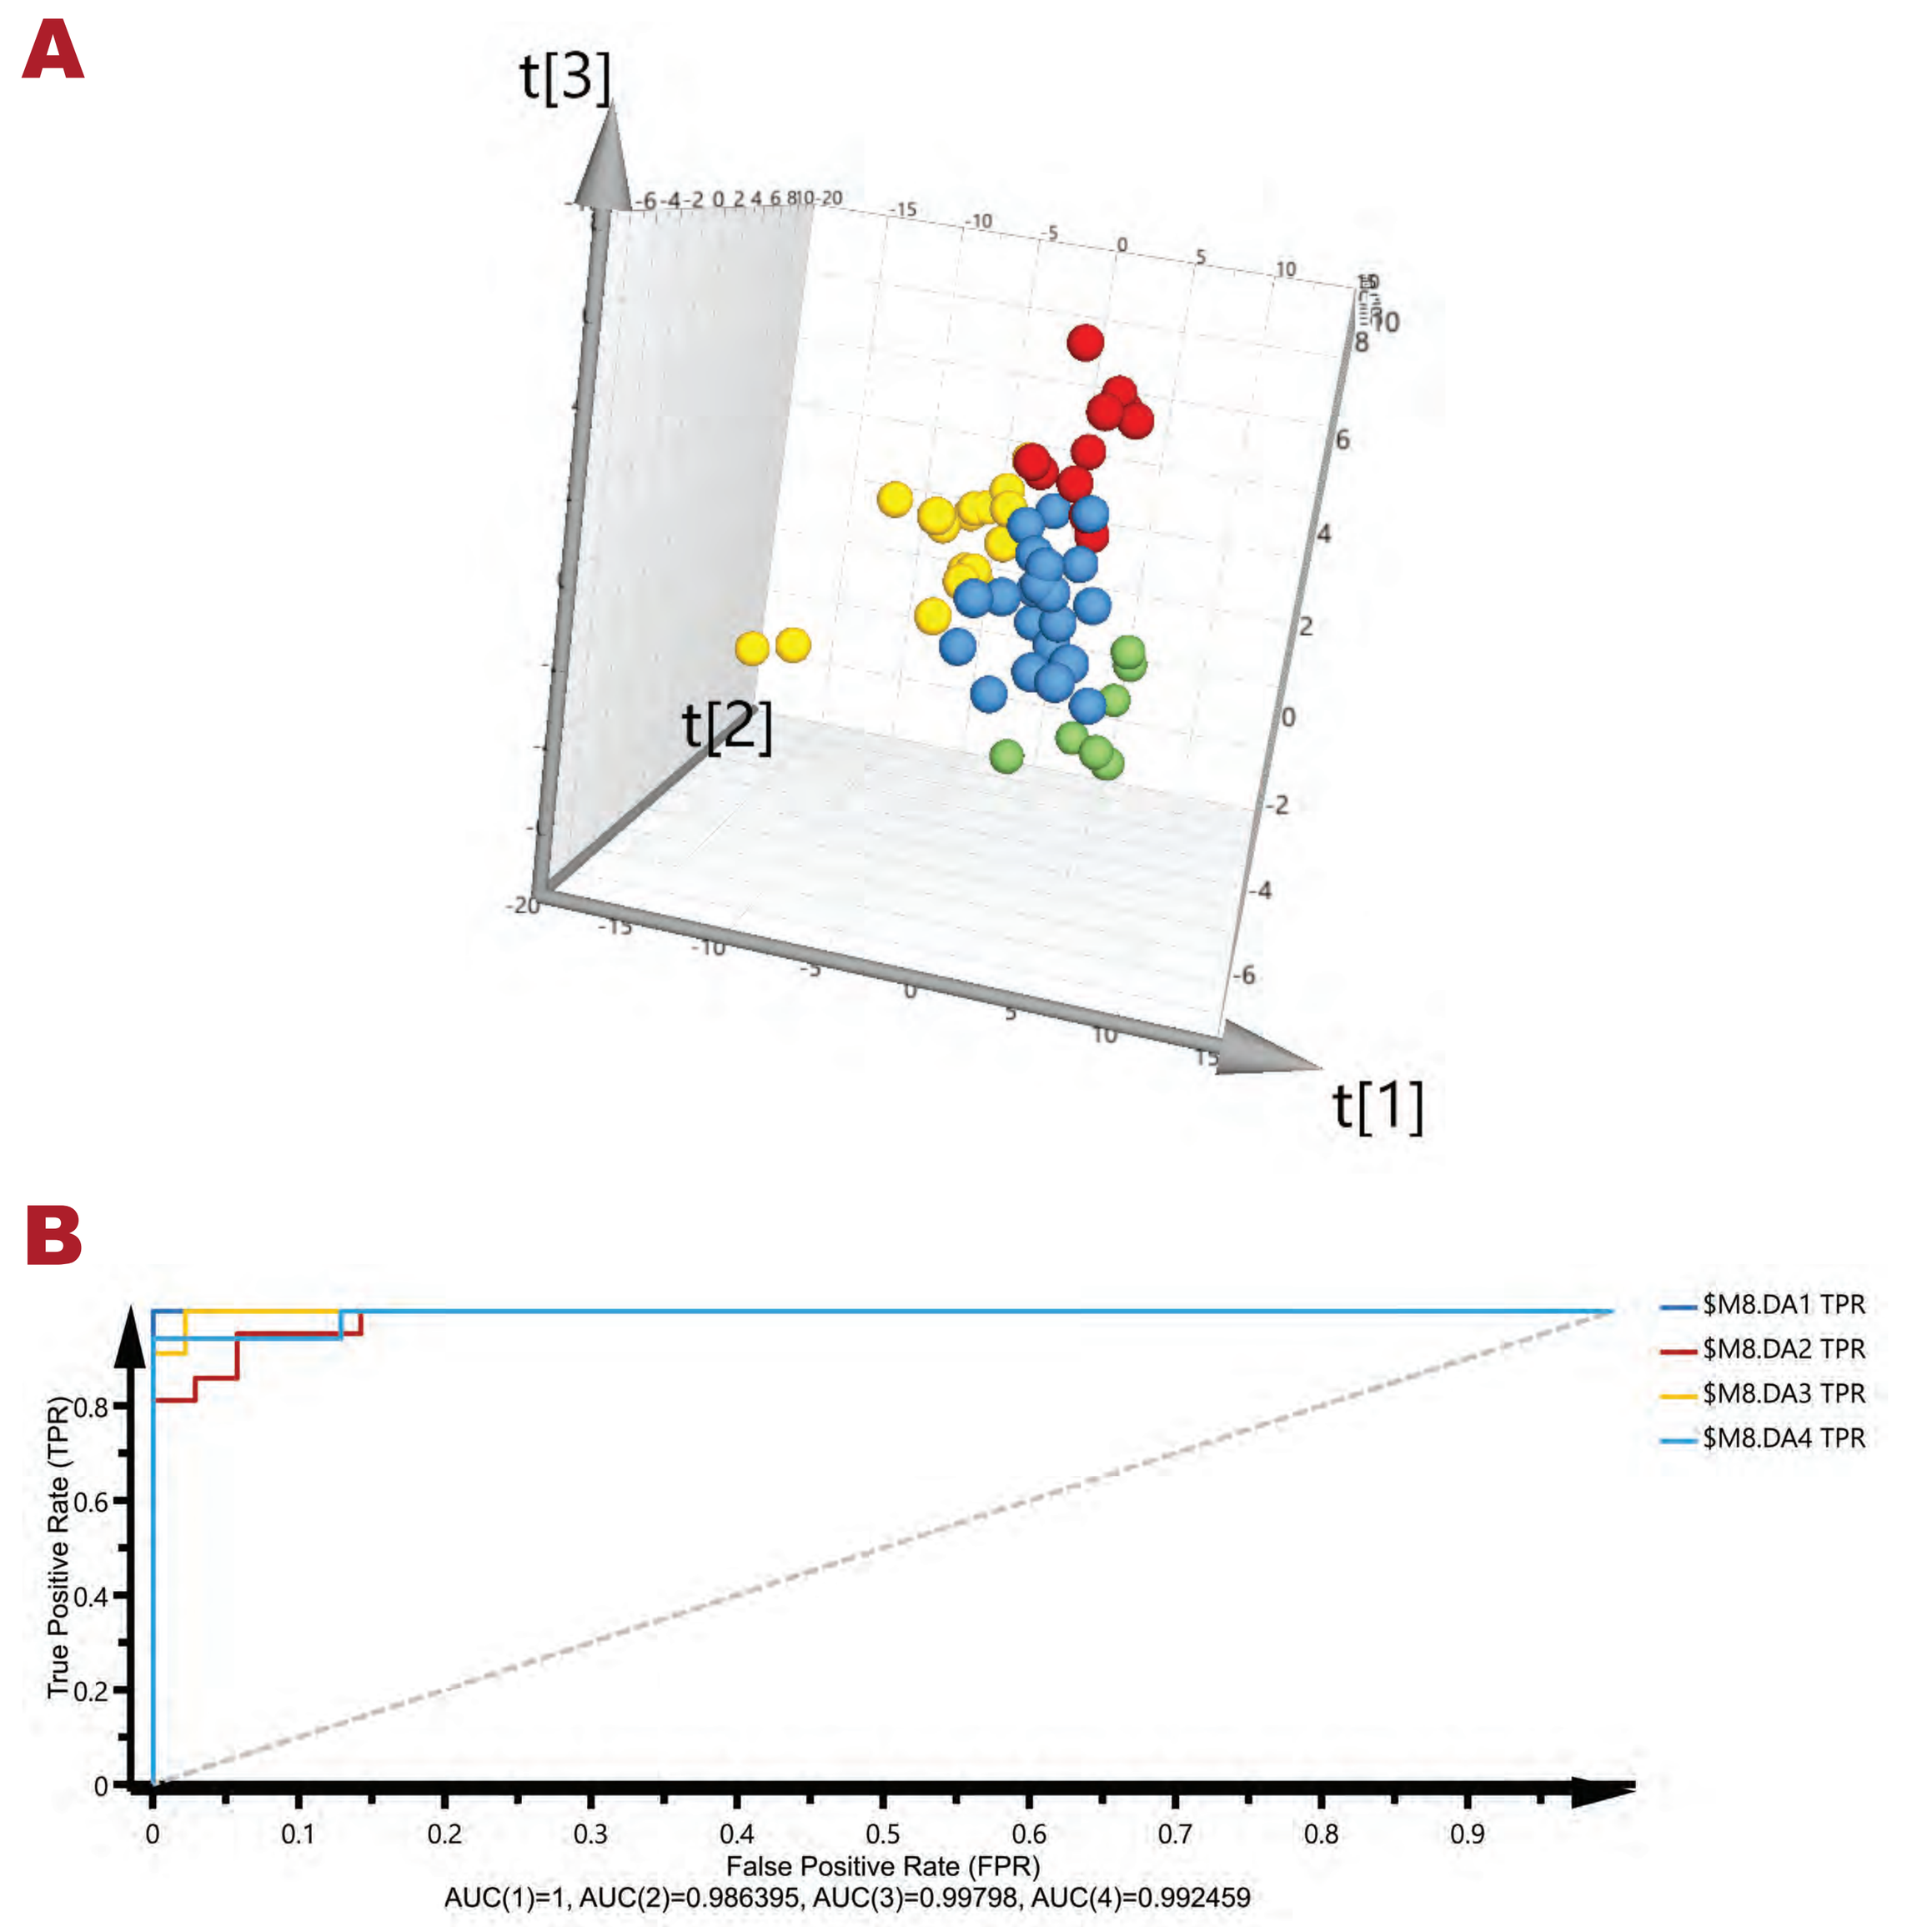
A
t[3]
t[2]
t[1]
B
True Positive Rate (TPR)
False Positive Rate (FPR)
AUC(1)=1, AUC(2)=0.986395, AUC(3)=0.99798, AUC(4)=0.992459
$M8.DA1 TPR
$M8.DA2 TPR
$M8.DA3 TPR
$M8.DA4 TPR

Figure 6. Simplification of the PCA model for clinical applications.
It is envisioned that the key proteins from the PCA model can be used further developed into diagnostic markers for subtyping PDAC. We first investigated the minimum number of proteins to maintain the specificity of the grouping in the multivariant analysis model. The 60 proteins with the most significant overall effect to the subtyping scheme were retained in the model. The score plot of the resulting analysis was shown in (A). The specificity of the sample grouping was accessed by the Receiver Operator Curve (ROC) in (B). DA1-4 signified Group1-4, respectively. ($R^2X = 0.633$; $R^2Y = 0.715$; $Q^2 = 0.566$)

PDAC Proteome versus Survival

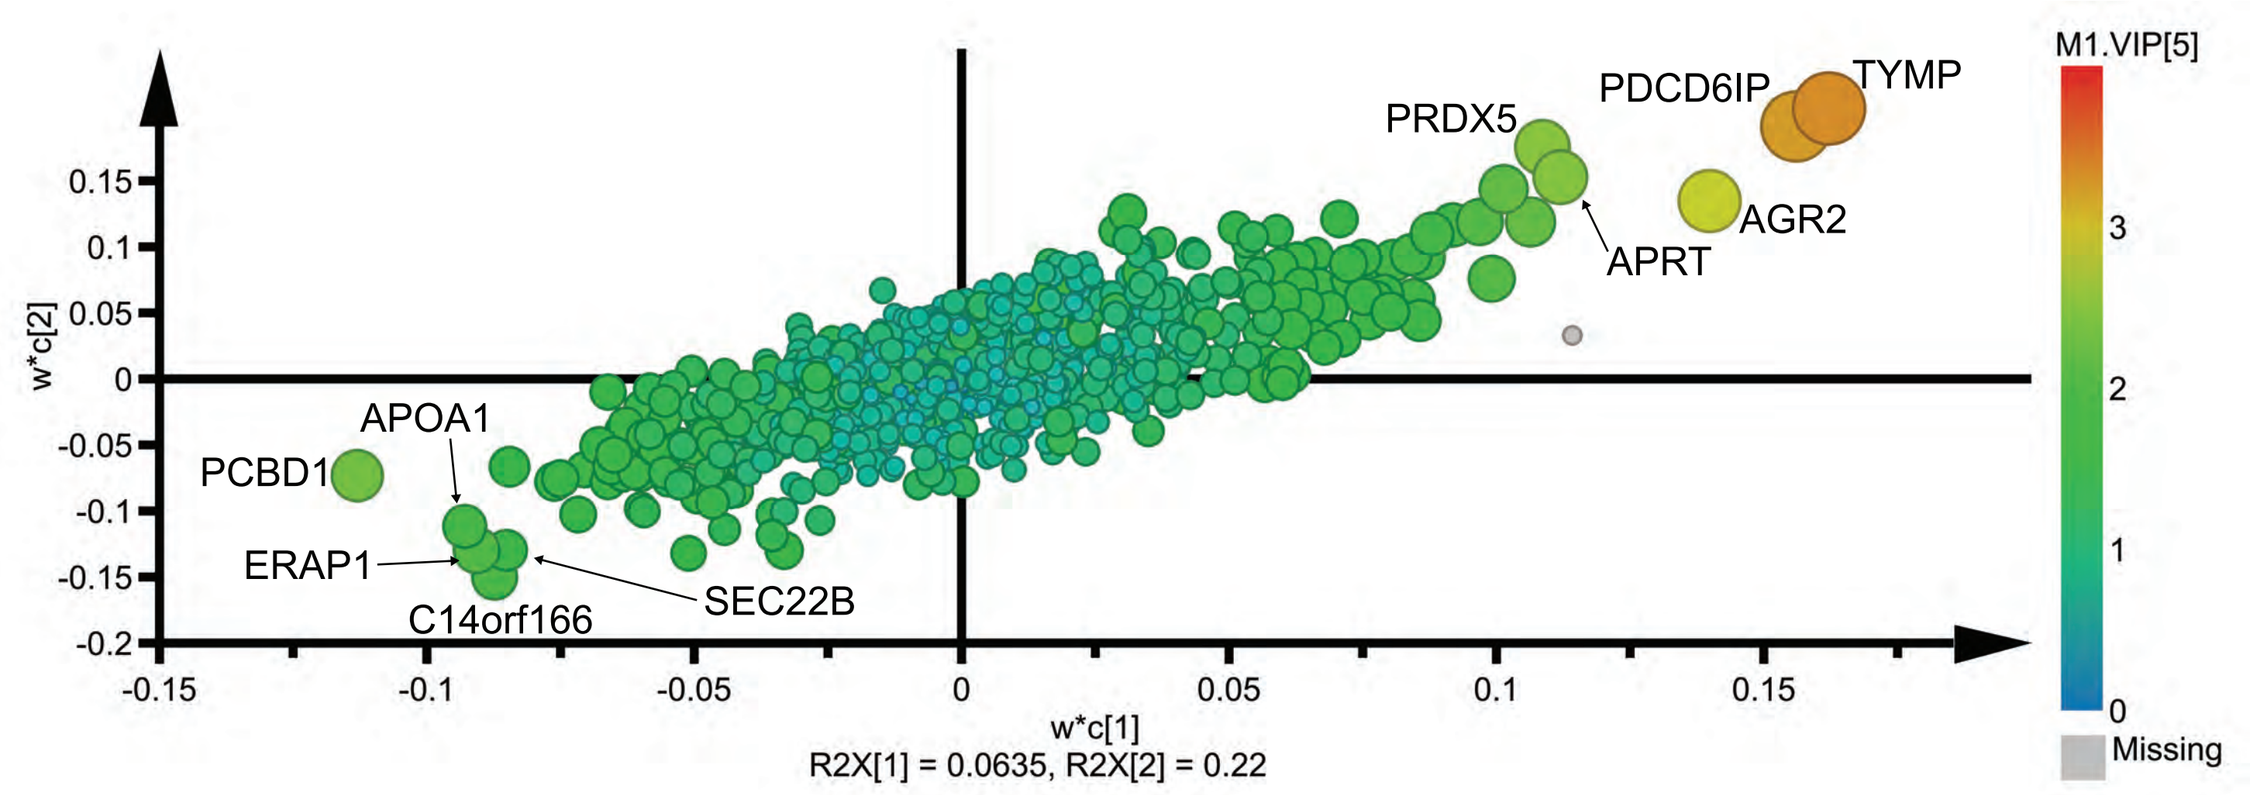
PRDX5
PDCD6IP
TYMP
AGR2
APRT
APOA1
PCBD1
ERAP1
C14orf166
SEC22B
w*c[1]
w*c[2]
R2X[1] = 0.0635, R2X[2] = 0.22
M1.VIP[5]
Missing

Figure 7. The loading plot of the partial least square analysis (PLS) analysis in the survival days versus the protein expressions.
This analysis was looking for biomarkers potentially correlating with the patient survival days after the diagnosis. In the loading plot, the color and size of the dots indicated the correlation between the protein and the survival days. Proteins on the right-hand side had a positive effect on the survival days and vice versa. The gene symbol of the proteins with the strongest influence to the model are labeled on the plot. ($R^2X(cum) = 0.477$; $R^2Y(cum) = 0.913$; $Q^2(cum) = 0.271$)

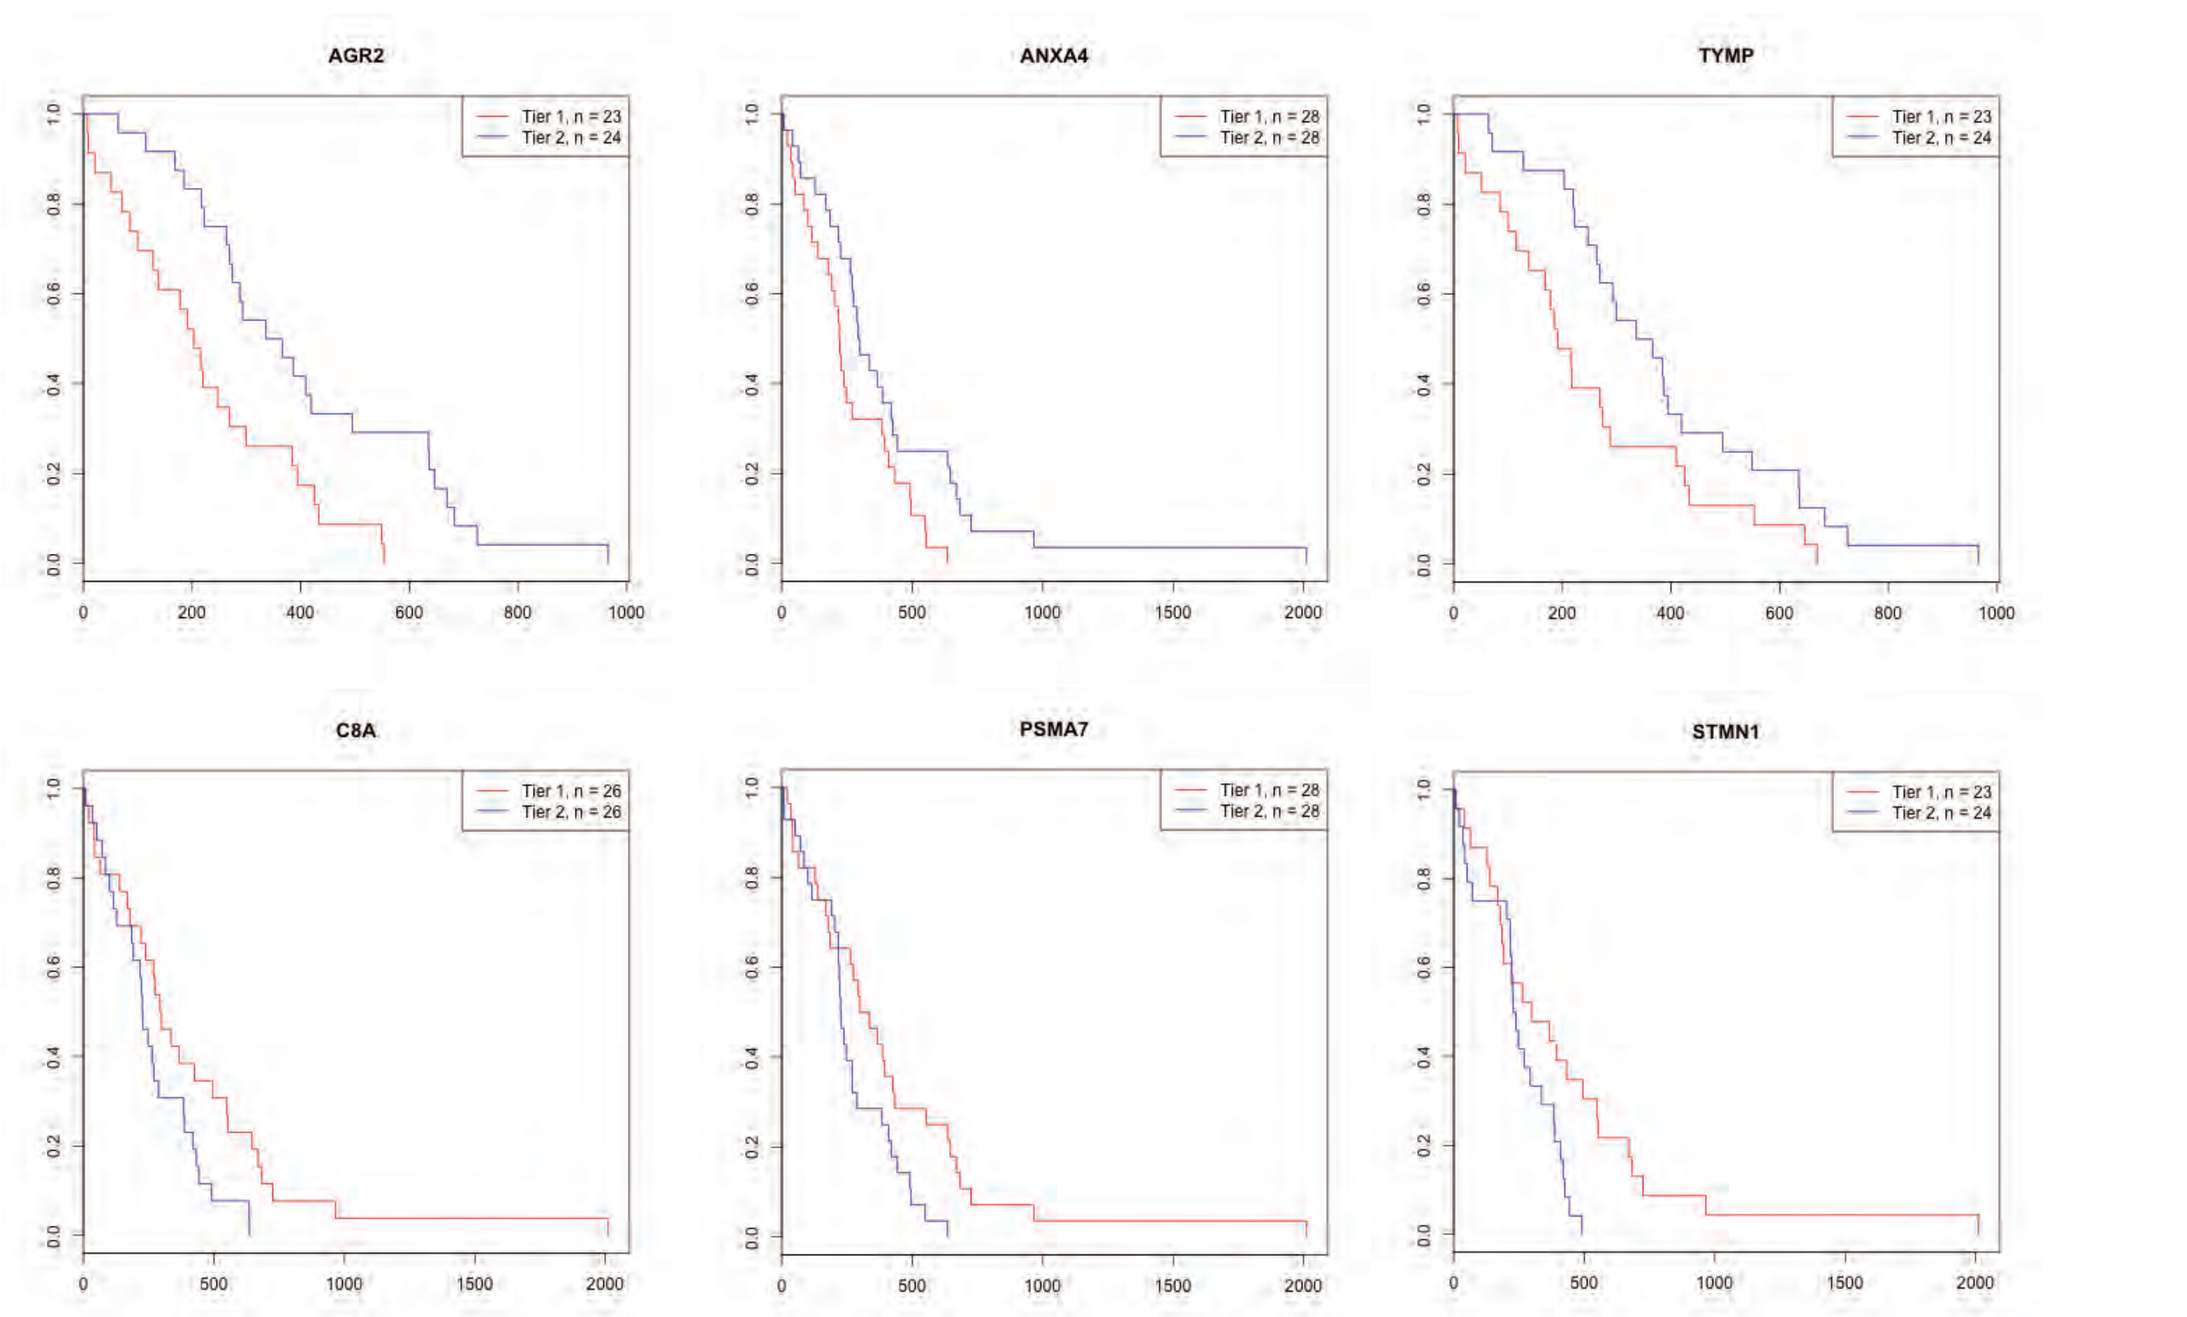
AGR2
ANXA4
TYMP
C8A
PSMA7
STMN1

Figure 8. Kaplan-Meier curves of 6 selected protein associated with patient survival
In this analysis, the patients were ranked according to the individual protein expression. Tier 1 and 2 represented the lower- and upper-half of the patient cohort. The lower-AGR2, ANXA4, TYMP, C8A, PSMA7 and STMN1 were some of the proteins found to be associated with the patient survival from the proteome (log-rank test, *p*-value <0.05).

Concluding remarks

The PCA analysis clustered the liver metastases samples into 4 groups. In the initial examination of the histology of the corresponding slides, there were significantly more stroma involvement in Group 4. The gene ontology analysis of the protein clusters suggested that there were stronger immune reponse in Group 4 than the rest of the samples. It was also found that there were significant associations with alcohol and tobacco usage in the PDAC subtype. The PDAC subtying scheme proposed herein was generally consistent with the genome-based classification system previously reported. The model could still provide accurate classification even when it was reduced to 60 proteins. In the PLS analysis, proteins correlating with survival days were identified. Thymidine phosphorylase (TYMP) was identified as one of the most influential proteins in the model. Patients with tumor overexpressing this protein had a better prognosis and had a better response to capecitabine treatment. Further analysis of the models may allow us to shed light on the mechanistic pathways and even prognostic biomarkers in pancreatic cancer.

Acknowledgement

This research was supported by the UNMC SPORE for Pancreatic Cancer Career Enhancement Program (CEP).

Reference

Bailey *et al*, Genomic analyses identify molecular subtypes of pancreatic cancer. Nature. 2016 Mar 3;531(7592):47-52.